#7006

MM-121: a human monoclonal antibody ErbB3 antagonist

Birgit Schoeberl, Kip A. West, Magda O. Leszczyniecka, Lin Nie, Olga Burenkova, Bryan E. Linggi, A.C. Fulgham, Joe Toth, Violette Paragas, Raghida Bukhalid, Viara Grantcharova , Matt Wallace, Lihui Xu, Washington Alves, Jose Varghese, Bill Slichenmyer, Michael J. Feldhaus, and Ulrik B. Nielsen

Merrimack Pharmaceuticals Inc., 700 One Kendall Sq, Cambridge, MA 02139

Abstract

The role of the EGFR family in cancer is well documented. To date most of the attention has been focused on EGFR and ErbB2. We have taken a systems approach to understanding ErbB signaling and built a comprehensive computational model of the pathway. Simulations revealed ErbB3 as an ultra-sensitive node in the ErbB pathway and suggested it as a better target for therapeutic intervention. Historically, ErbB3 has not been considered as an attractive target because of its impaired kinase activity and low expression in tumors although recent publications suggest a key role in promoting resistance to EGFR and ErbB2 targeted therapeutics.

Human monoclonal IgG2 antibodies against ErbB3 were then developed and tested in multiple orthogonal assays for the ability to modulate ErbB3 activity and one candidate, MM-121, was chosen to move forward into pre-clinical testing. MM-121 demonstrated potent inhibition of heregulin-induced signaling events in human cancer cell lines.

When MM-121 efficacy was assessed in vivo, MM-121 showed a dose-dependent ability to inhibit tumor growth in multiple xenograft models of human cancer, including ovarian, renal cell, pancreatic and prostate cancer. Pharmacodynamic analysis of samples from these studies showed that chronic MM-121 administration resulted in inhibition of ErbB3 activity which correlated with efficacy.

In vitro antibody binding studies suggested that MM-121 binds to ErbB3 receptors from mouse, rat, cynomolgus monkey and human with a similar affinity. In order to design an efficacious and safe dosing regimen, pre-clinical studies were carried out in mouse, rat and cynomolgus monkey. These studies suggest that MM-121 can safely be administered at serum levels that produce in good inhibition of ErbB signaling and efficacy in xenograft models of cancer.

Cumulatively, these data support that MM-121 could have significant clinical benefit for the treatment of cancer and warrant proceeding forward into phase 1 clinical trials.

Mathematical Model of the ErbB Pathway

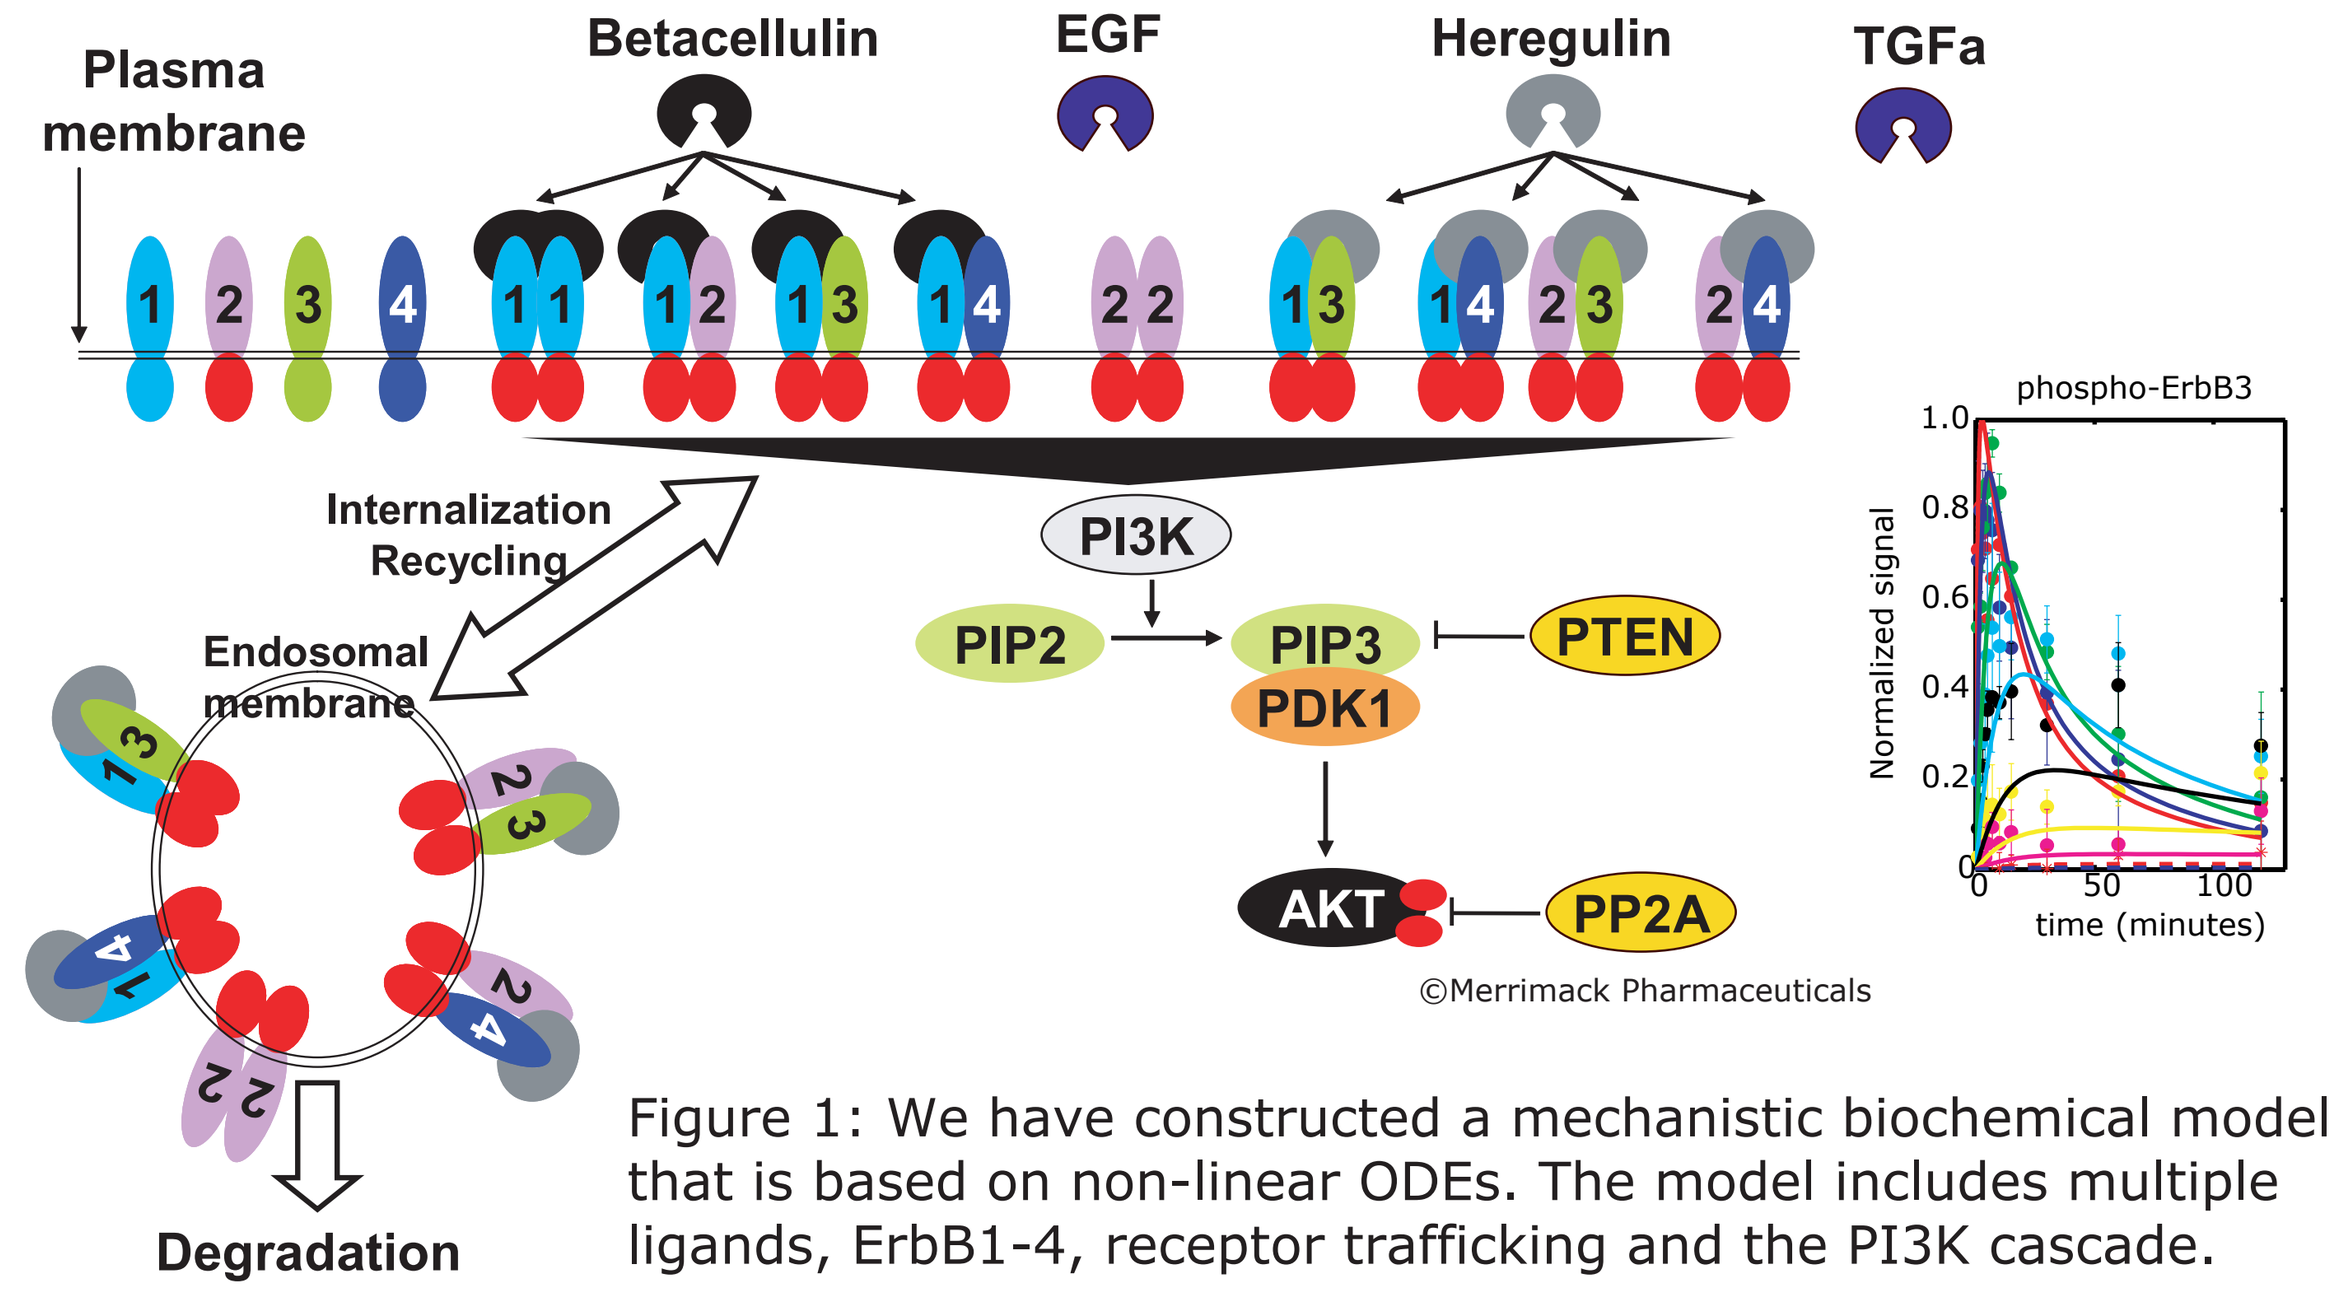
Plasma membrane
Betacellulin
EGF
Heregulin
TGFa
Internalization Recycling
Endosomal membrane
Degradation
PI3K
PIP2
PIP3
PDK1
PTEN
AKT
PP2A
phospho-ErbB3
Normalized signal
time (minutes)
©Merrimack Pharmaceuticals

Figure 1: We have constructed a mechanistic biochemical model that is based on non-linear ODEs. The model includes multiple ligands, ErbB1-4, receptor trafficking and the PI3K cascade.

Sensitivity analysis identified target ErbB3

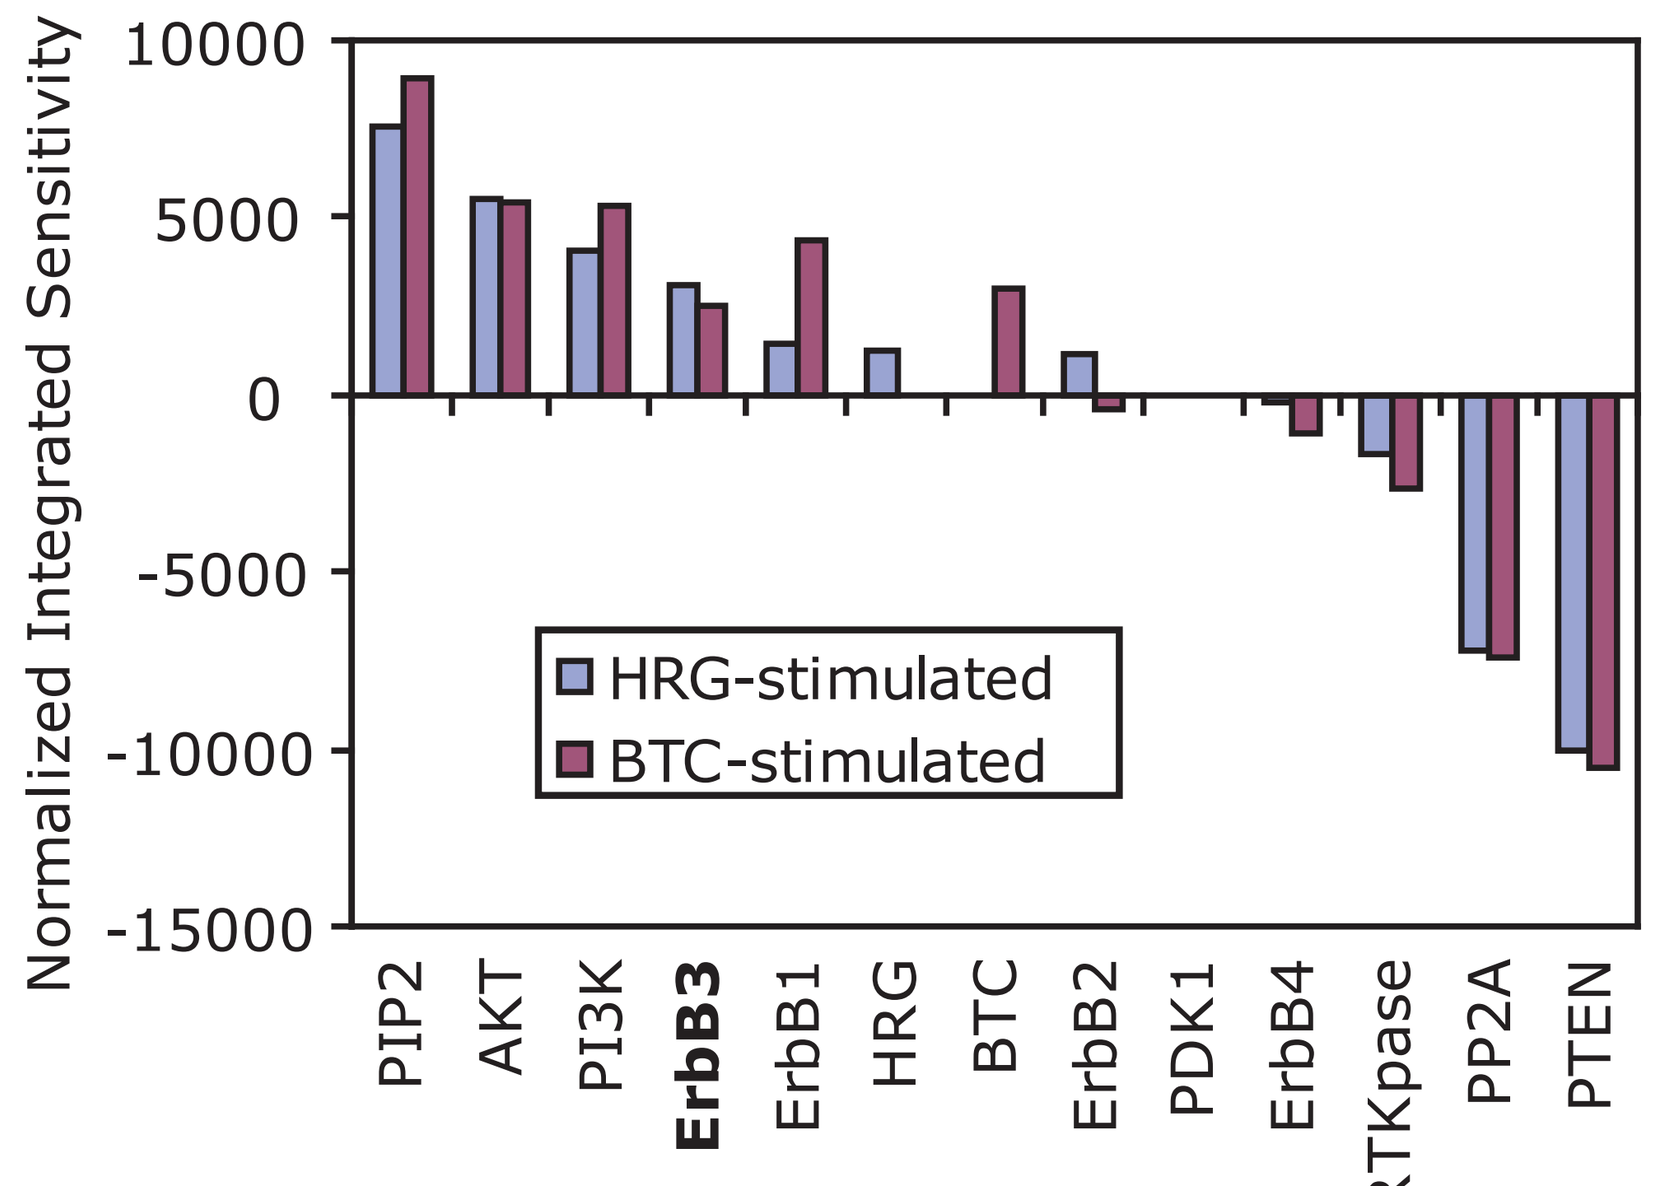
Normalized Integrated Sensitivity
HRG-stimulated
BTC-stimulated
PIP2
AKT
PI3K
ErbB3
ErbB1
HRG
BTC
ErbB2
PDK1
ErbB4
RTKpase
PP2A
PTEN

Figure 2: Mass action kinetic equations were used to create a model in MATLAB SimBiology v2.1. This model contains >60 species, >50 reactions, and ~14 parameters. Kinetic parameters reported in the literature were if used available; however, sensitivity analysis and parameter estimatio been performed.

MM-121 inhibits HRG induced signaling in ADRr and ACHN cells

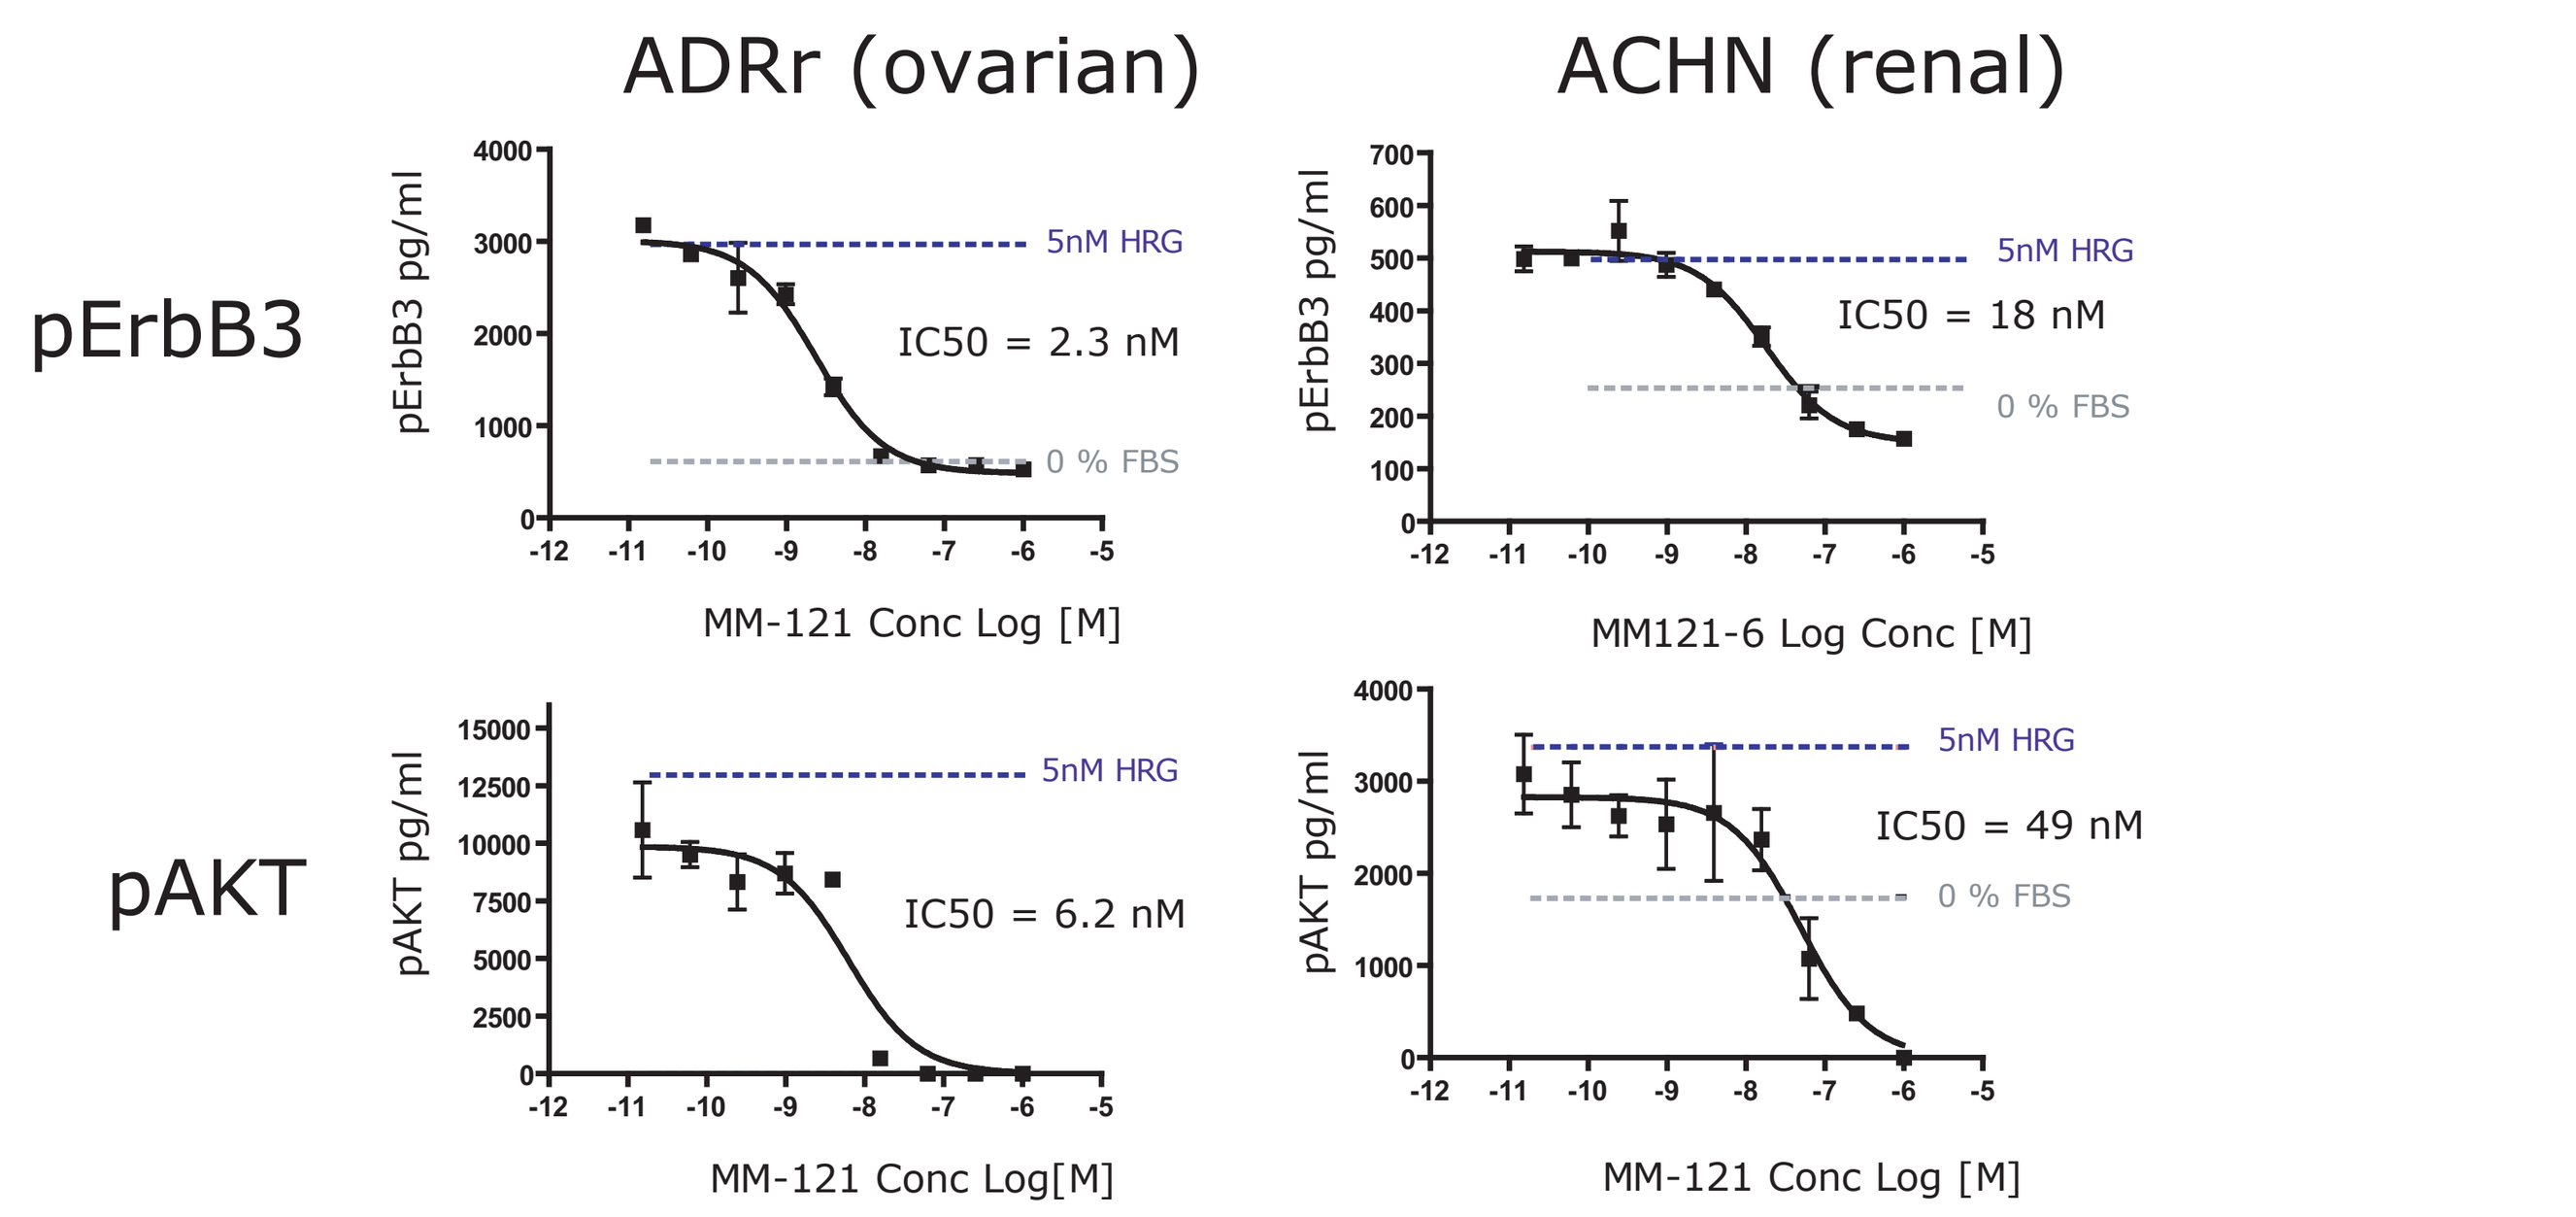
ADRr (ovarian)
ACHN (renal)
pErbB3
pAKT
pErbB3 pg/ml
pAKT pg/ml
5nM HRG
0 % FBS
IC50 = 2.3 nM
IC50 = 18 nM
IC50 = 6.2 nM
IC50 = 49 nM
MM-121 Conc Log [M]
MM121-6 Log Conc [M]
MM-121 Conc Log[M]
MM-121 Conc Log [M]

Figure 3: MM-121 inhibition of ErbB3 and AKT phosphorylation in serum starved ADRr and ACHN cells stimulated with 5 nM HRG1β - 1 *in vitro*. pErbB3 and pAKT were measured by ELISA.

MM-121 inhibits migration in a transwell assay

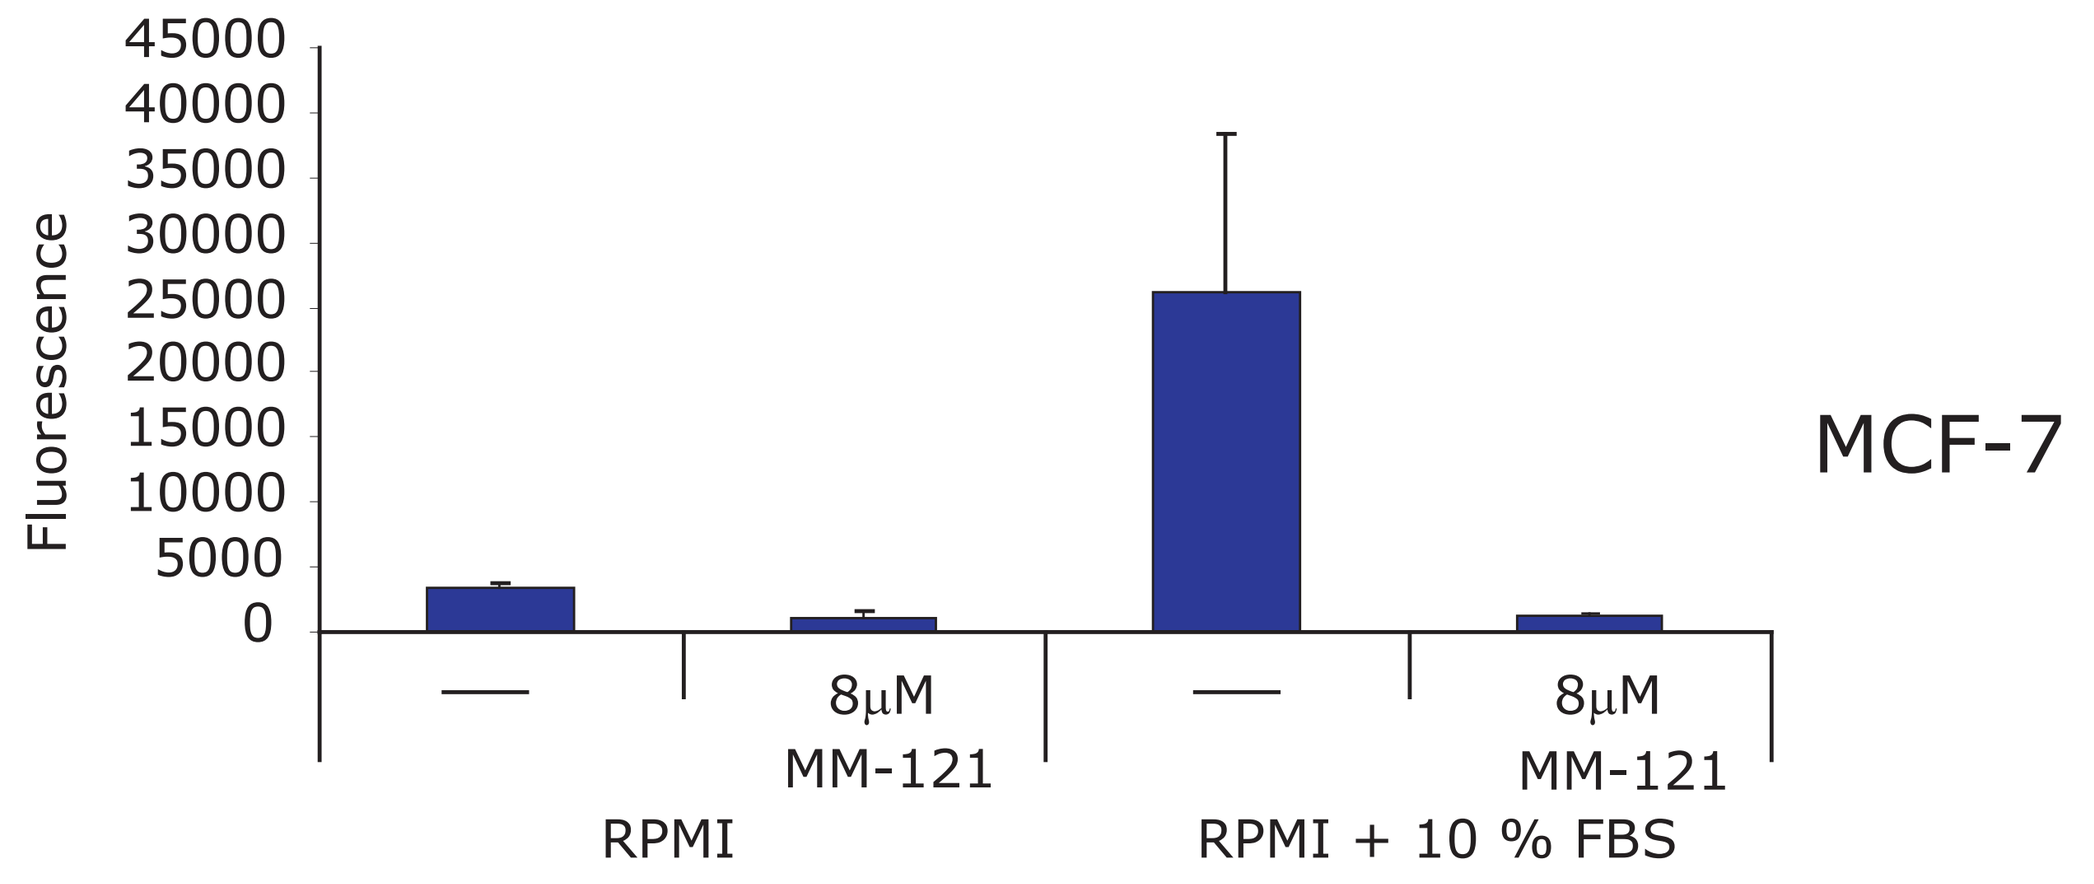
Fluorescence
MCF-7
8μM MM-121
8μM MM-121
RPMI
RPMI + 10 % FBS

Figure 4: MM-121 inhibits cell motility in a transwell assay. MCF-7 cells were incubated for 15 minutes in 8 M MM-121 and then placed in the top chamber of a transwell and migration towards serum-free media or media supplemented with 10% FBS, in the presence or absence of MM-121 (8μM), was assessed after 16 hrs.

MM-121 inhibits spheroid growth

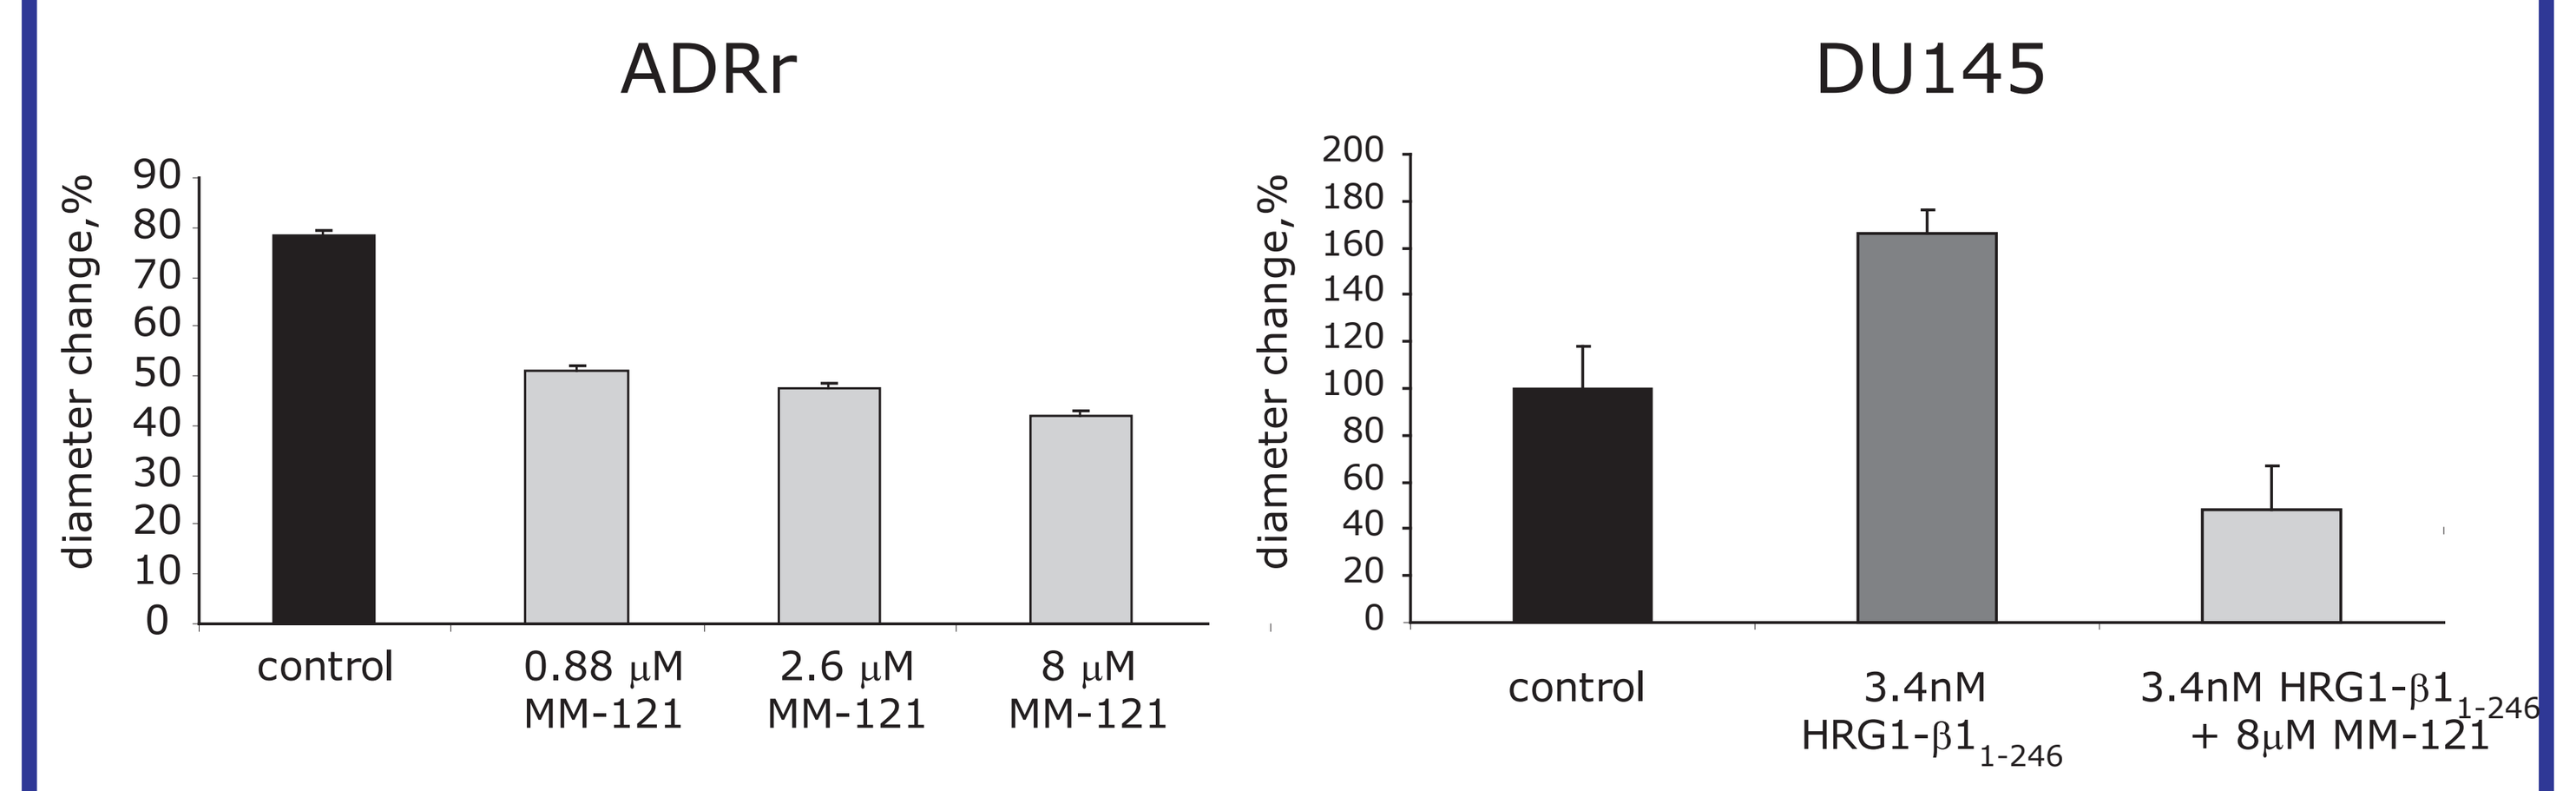
ADRr
DU145
diameter change,%
control
0.88 μM MM-121
2.6 μM MM-121
8 μM MM-121
control
3.4nM HRG1-β1 1-246
3.4nM HRG1-β1 1-246 + 8μM MM-121

Figure 5: MM-121 inhibits spheroid growth in the presence or absence of HRG1- $\beta 1_{1\text{-}246}$. (A) MM-121 inhibits spheroid growth in ADRr cells in the absence of HRG1- 11-246 (B) HRG1- $\beta 1_{1\text{-}246}$ induces spheroid growth in DU145 cells which can be inhibited by MM-121.

MM-121 inhibits phospho-ErbB3 in PD studies

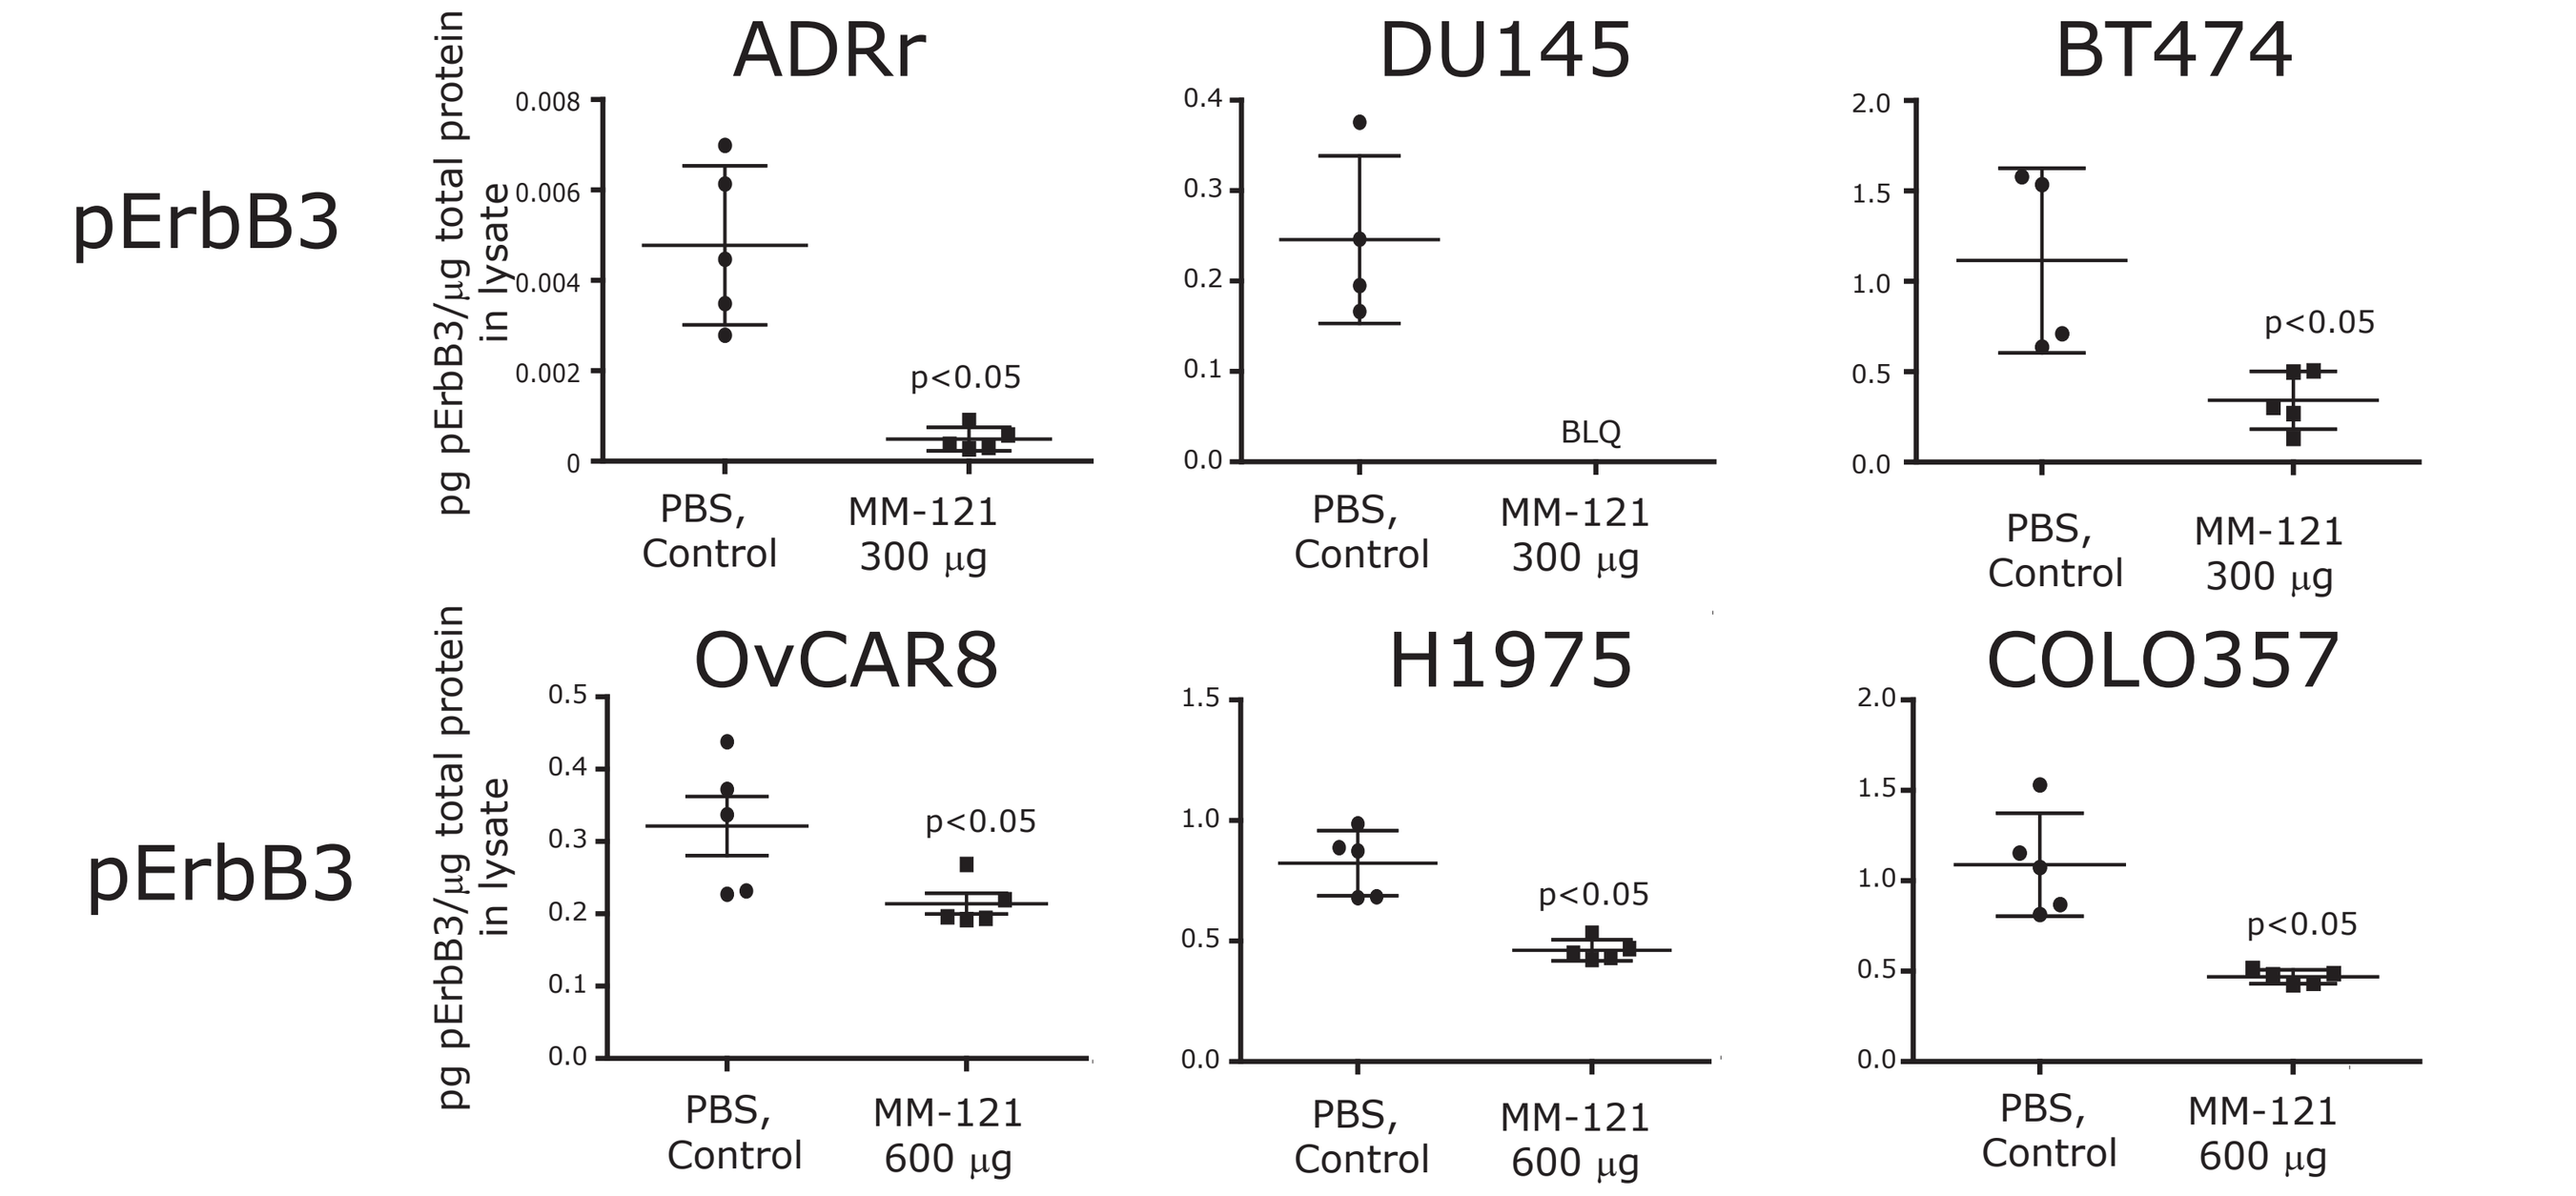
ADRr
DU145
BT474
OvCAR8
H1975
COLO357
pErbB3
pg pErbB3/μg total protein in lysate
PBS, Control
MM-121 300 μg
MM-121 600 μg
p<0.05
BLQ

Figure 6: Summary of pharmacodynamic studies assessing the inhibition of ErbB3 phosphorylation (pErbB3) by MM-121 levels in vivo. Tumor bearing mice were injected twice with 300 μg/inj or 600 μg/inj MM-121 every three days. Tumors were harvested 24 hours post last dose after the second dose. pErbB3 levels was measured in tumor lysate and normalized to total protein levels. pErbB3 levels are presented for the single animals. BLQ (Below Limit of Quantitation)

MM-121 inhibits VEGF secretion *in vitro* and *in vivo*

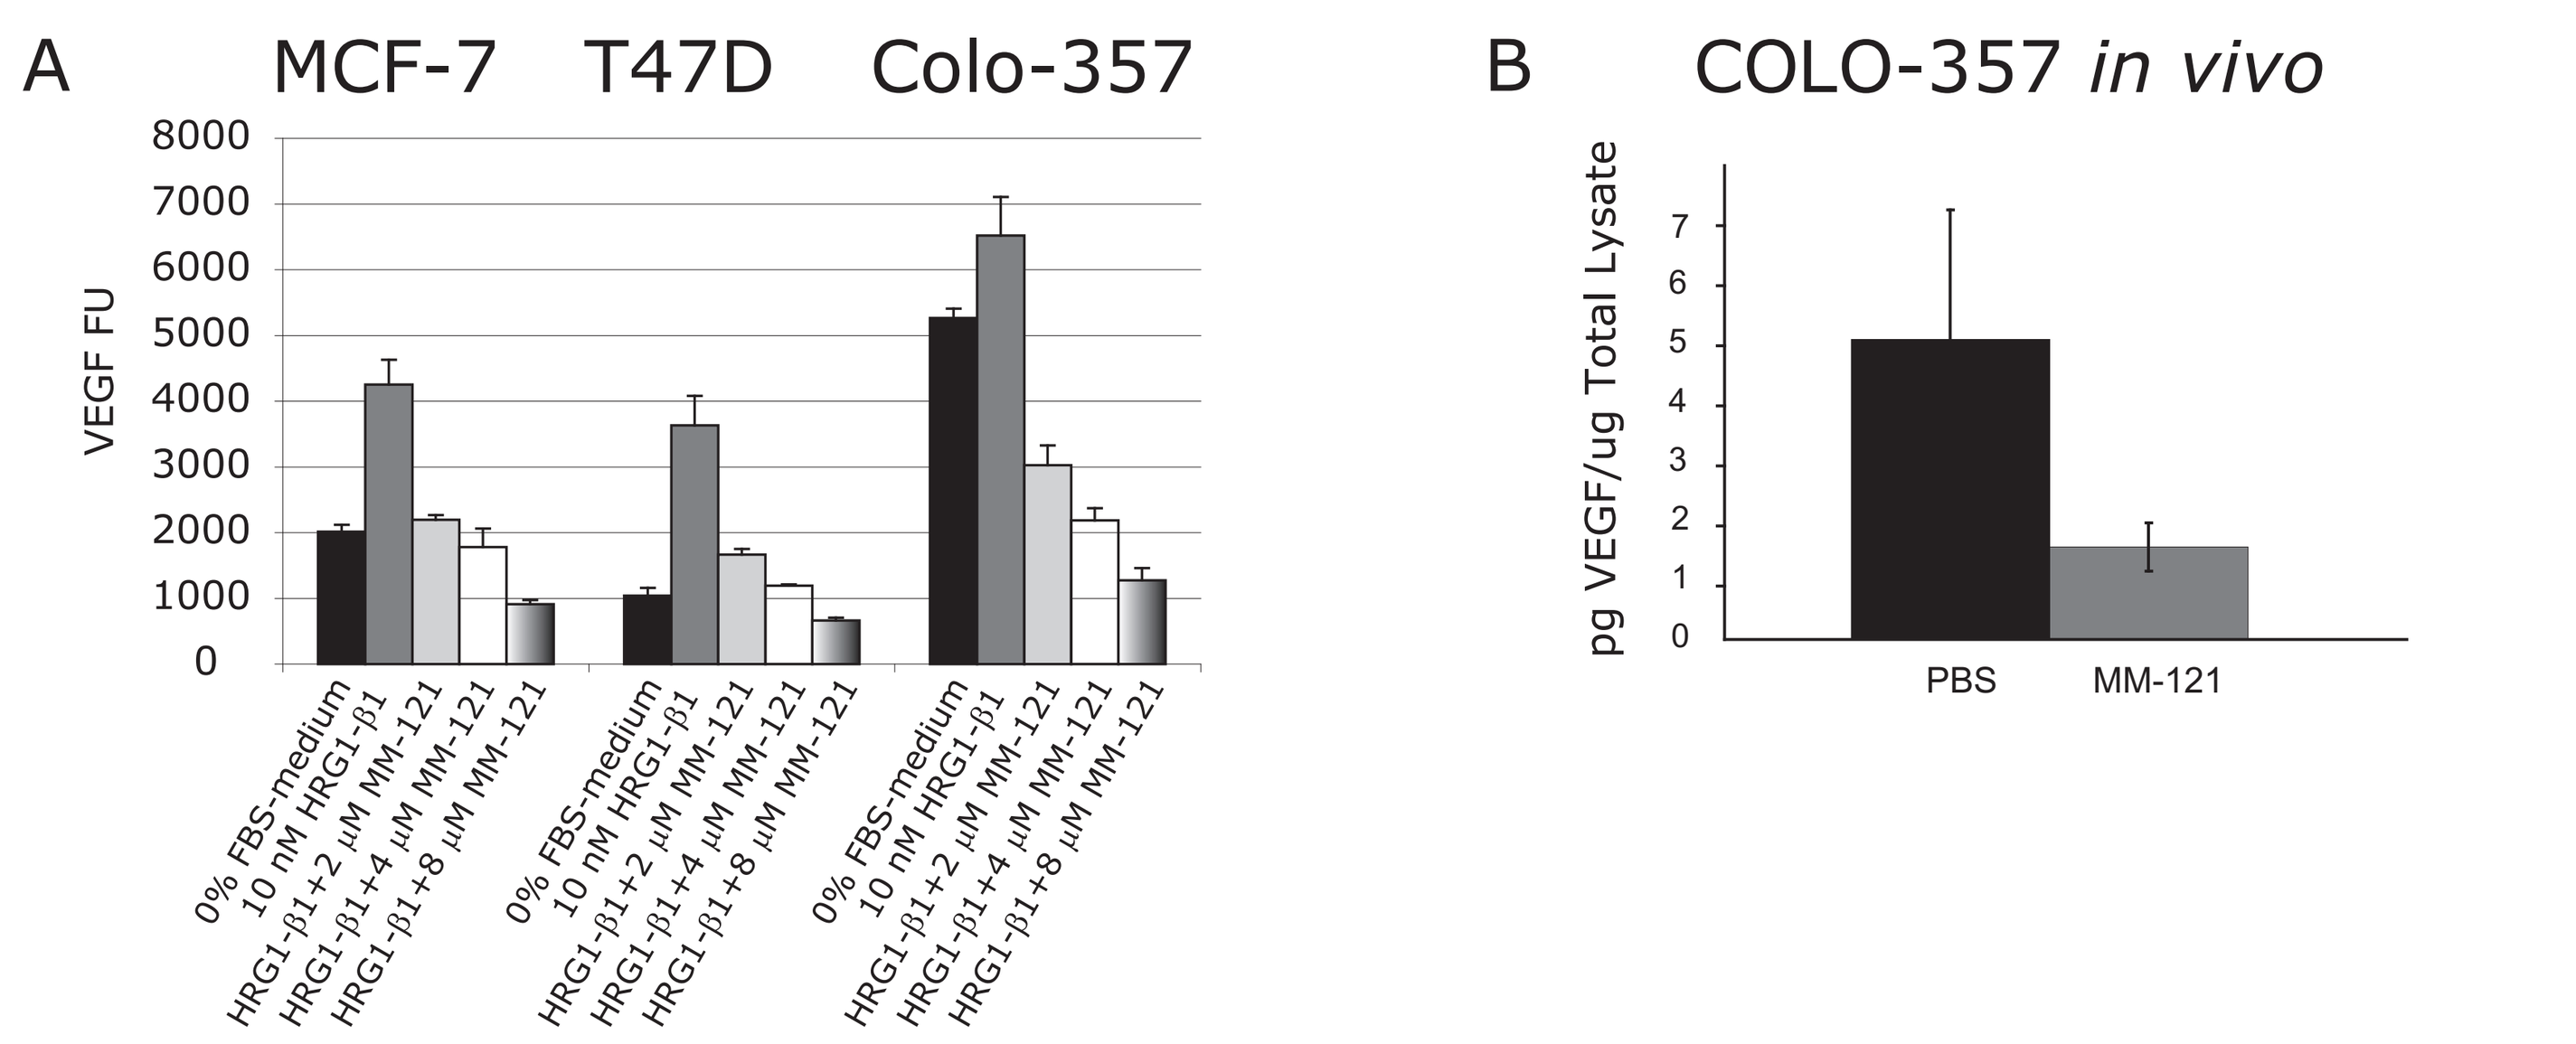
A
MCF-7
T47D
Colo-357
VEGF FU
0% FBS-medium
10 nM HRG1-β1
HRG1-β1+2 μM MM-121
HRG1-β1+4 μM MM-121
HRG1-β1+8 μM MM-121
B
COLO-357 in vivo
pg VEGF/ug Total Lysate
PBS
MM-121

Figure 7: MM-121 inhibits in vitro and in vivo secretion of VEGF. (A) MM-121 inhibits VEGF secretion in serum starved MCF-7, T47D and COLO357 cells cultured in 10 nM HRG1-β1 in the presence of increasing amounts of MM-121 (2 μM, 4 μM and 8 μM) for 48 hrs. VEGF secretion was measured by ELISA. (B) The efficacy of MM-121 in the in vivo inhibition of VEGF production was assessed in COLO357 (pancreatic). Tumors were established in nude mice and animals were dosed intra-peritoneally q3d for 1 week with a 600 μg/mouse concentration of MM-121. 24 hours post final dose tumors were harvested and analyzed for intra-tumoral levels of VEGF by ELISA .

MM-121 toxicology experience

- MM-121 was well-tolerated in cynomolgus monkeys when administered once weekly for nine weeks at 8, 32 and 160mg/kg and in Sprague-Dawley rats when administered twice a week at 5, 25 and 100mg/kg

- The only MM-121-related effect of toxicological significance was an increase in spleen organ weight in the 160mg/kg dose group of the monkey study

MM-121 efficacy *in vivo*

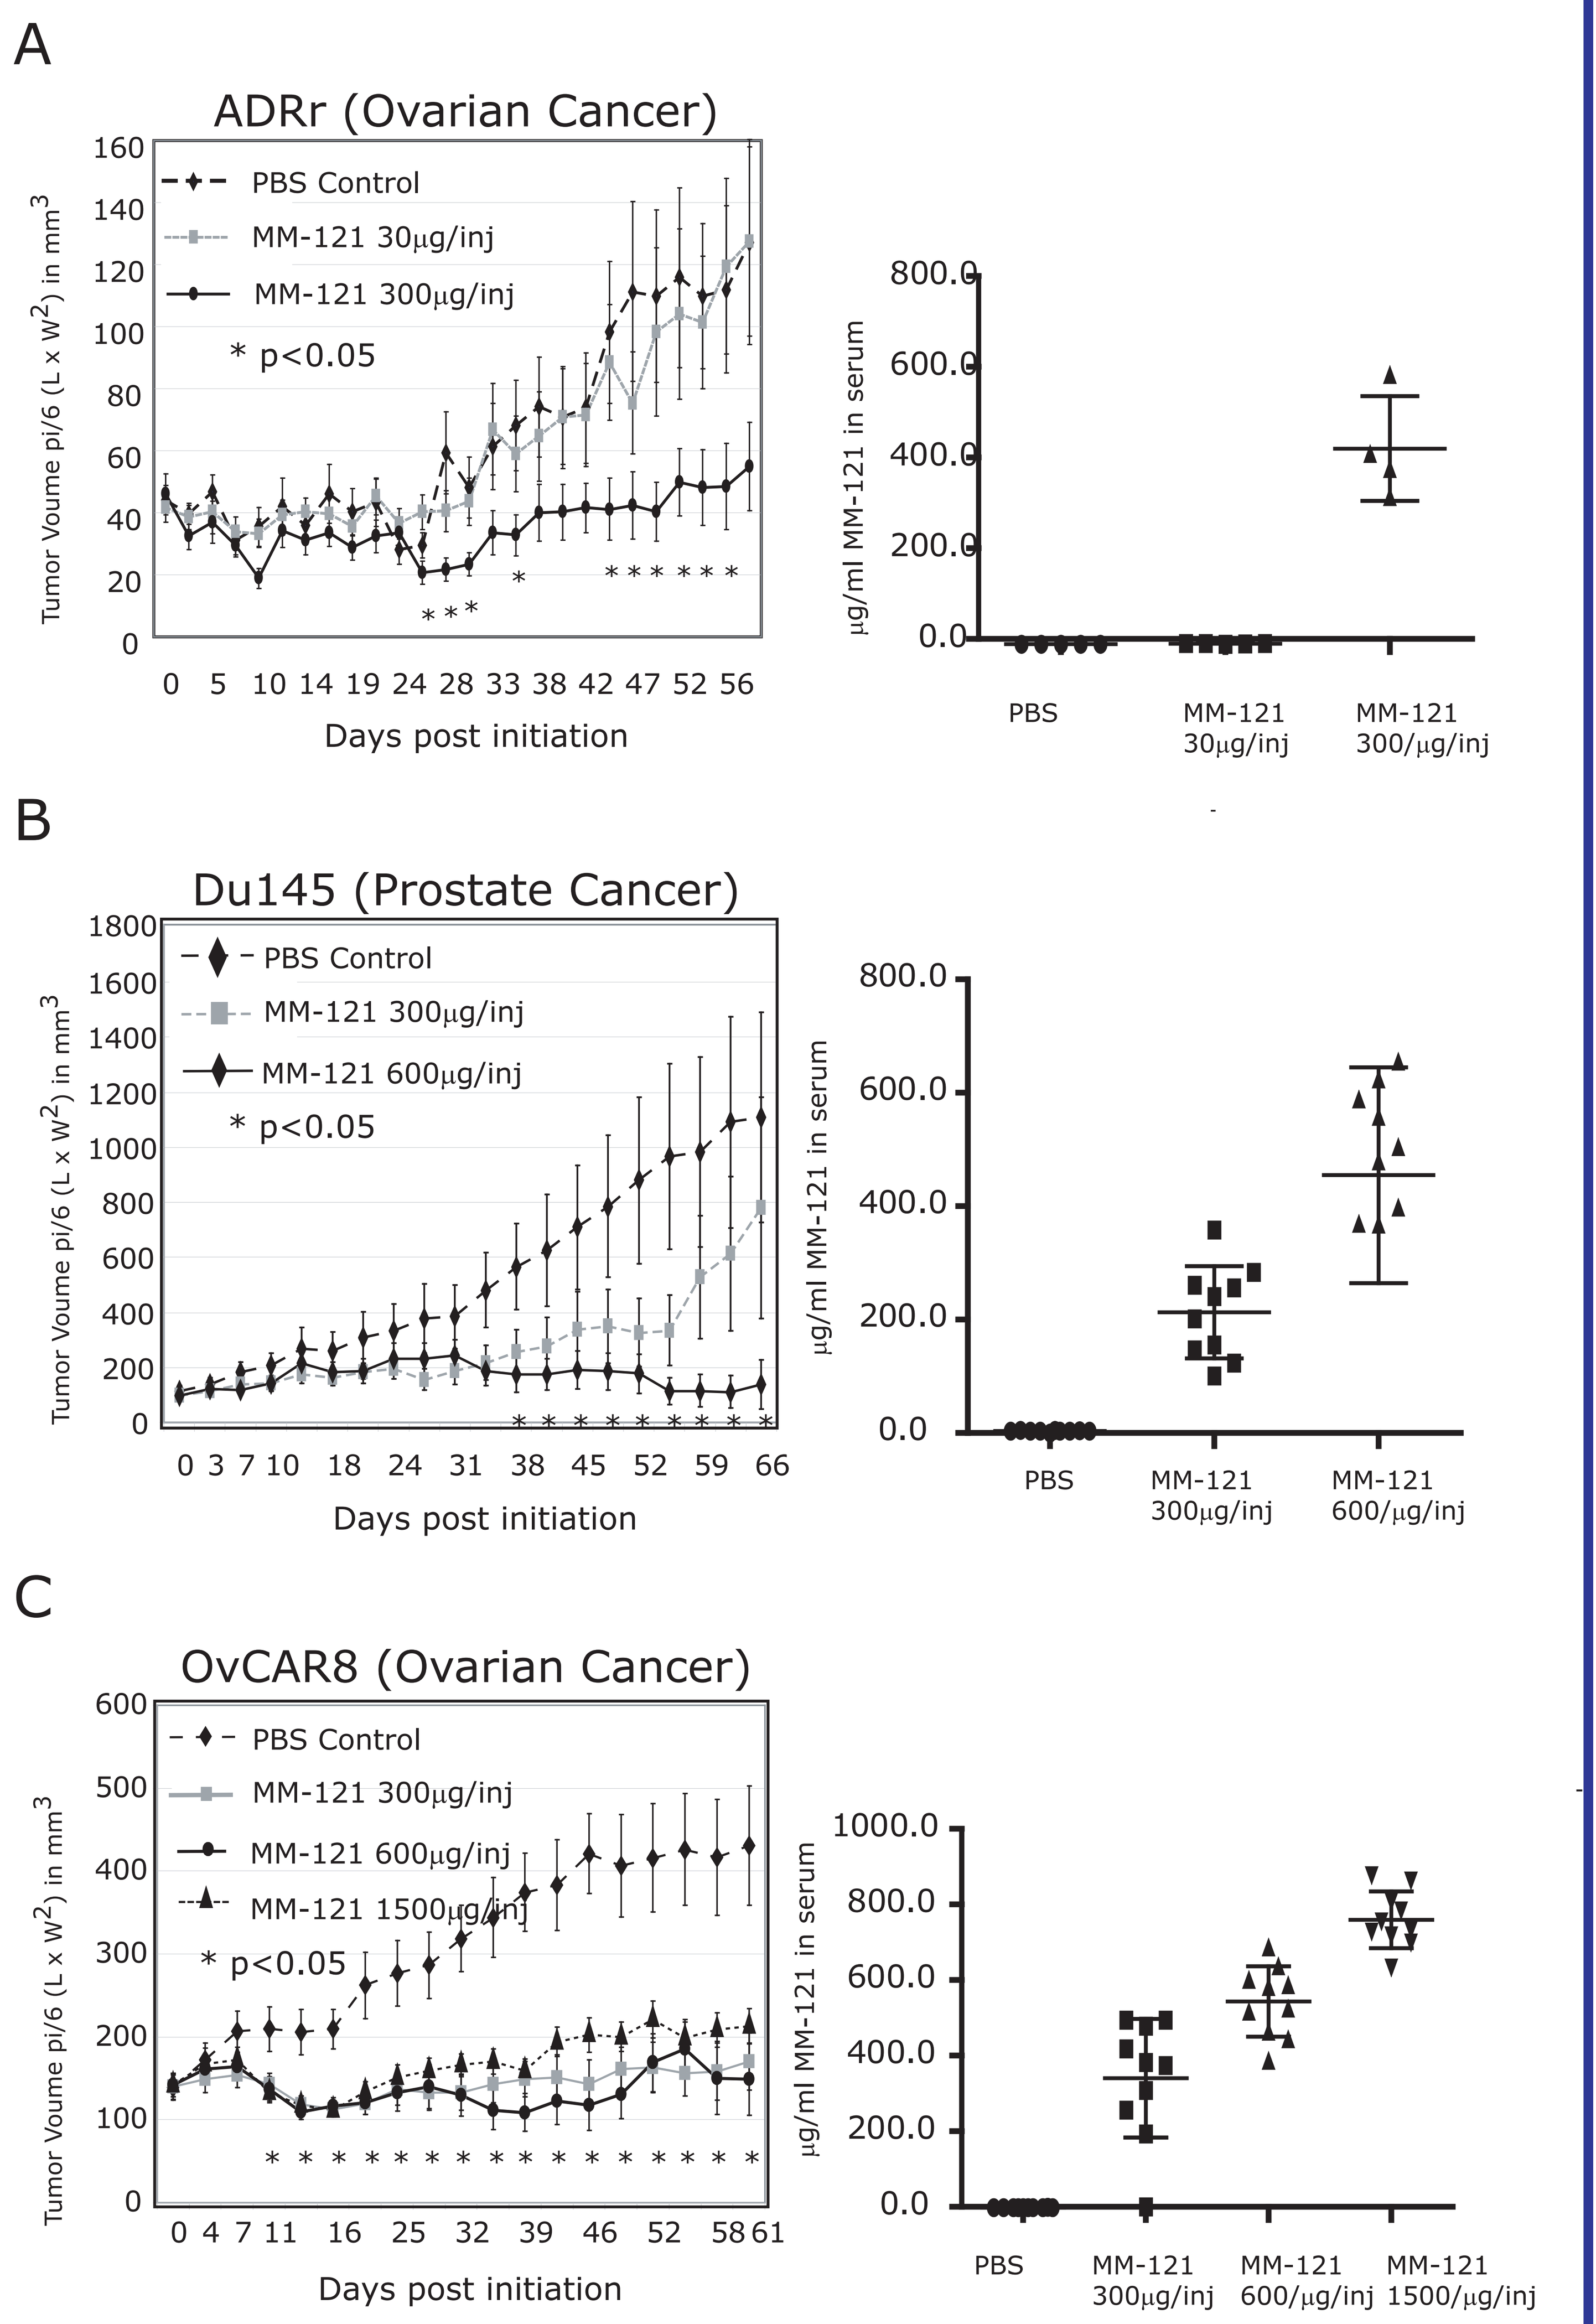
A
ADRr (Ovarian Cancer)
PBS Control
MM-121 30μg/inj
MM-121 300μg/inj
* p<0.05
Tumor Voume pi/6 (L x W2) in mm3
Days post initiation
μg/ml MM-121 in serum
PBS
MM-121 30μg/inj
MM-121 300/μg/inj
B
Du145 (Prostate Cancer)
PBS Control
MM-121 300μg/inj
MM-121 600μg/inj
MM-121 300μg/inj
MM-121 600/μg/inj
C
OvCAR8 (Ovarian Cancer)
PBS Control
MM-121 300μg/inj
MM-121 600μg/inj
MM-121 1500μg/inj
MM-121 600/μg/inj
MM-121 1500/μg/inj

Figure 8: MM-121 Efficacy in xenograft models of human cancer-The efficacy of MM-121 in the inhibition of tumor growth was assessed at multiple doses in 3 xenograft cancer models derived from human tumor cell lines. (A) ADRr (ovarian), (B) Du145 (prostate) and (C) OvCAR8 (ovarian). Tumors were established in nude mice and animals were dosed intra-peritoneally q3d with different concentrations of MM-121. Tumor size (Length x Width) was measured twice a week and measurements were used to calculate tumor volume (π/6 (L x W2). * = p<0.05. MM-121 serum levels were measured 24 hours post last dose. The serum levels for the single animals and standard deviations are graphed.